A BIOMARKER-BASED HUMAN STEM CELL ASSAY APPLIED FOR RANKING A RETINOID SERIES BASED ON RELATIVE DEVELOPMENTAL TOXICITY POTENTIAL

Egnash LA, Palmer JA, Smith AM, Conard KR, Burrier RE, Donley ELR, Kirchner, FR

Stemina Biomarker Discovery Inc., 504 S. Rosa Rd., Suite 150, Madison WI 53719

www.stemina.com

Introduction

We previously developed an in vitro, biomarker-based, human induced pluripotent stem (iPS) cell-based assay to screen compounds for developmental toxicity. The assay measures changes in two amino acids (ornithine and cystine) involved in cell proliferation and differentiation. This assay (devTOX^{qP}) is currently being applied as an alternative model to aid in ongoing worldwide efforts to reduce animal testing.

In this work we demonstrate the use of the assay to rank order the relative developmental toxicity potential of compounds within a chemical series using a well-characterized set of retinoids. Results were compared to published data for both *in vivo* and other *in vitro* models. *In vivo* response to retinoids is variable for both human and animal models due to the developmental toxicity potency of these compounds in early development. Because retinoic acid signaling plays a key role in embryogenesis we also tested the mechanistic relevance of our *in vitro* model using the retinoic acid receptor (RAR) antagonist Ro 41-5253. Overall, rankings in the absence of the antagonist were concordant with published values for other *in vitro* studies, but do not completely correspond with *in vivo* potency rankings. The lack of *in vivo* kinetic and metabolic processes and species-specific differences in metabolism could explain these differences. Observations in the presence of Ro 41-5253 were consistent with the developmental toxicity response being mediated through RAR and suggest iPS cells do not have the ability to metabolize etretinate to its active form. These data illustrate the importance of the RAR pathway in mediating developmental toxicity responses and show how this assay can be applied for compound decision making and bridging chemical series.

Methods

Assay Workflow

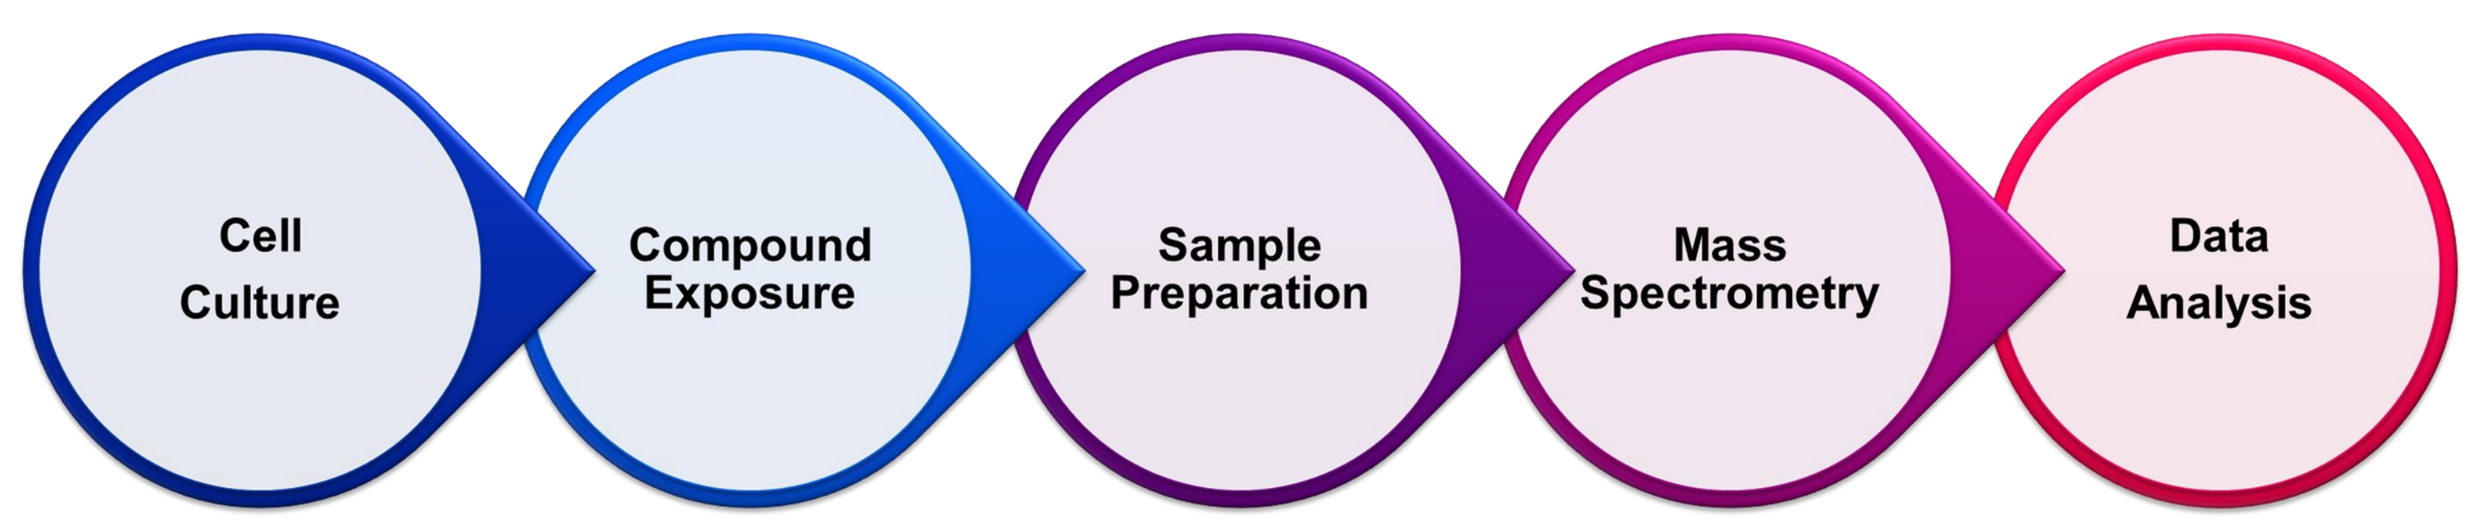
Cell Culture
Compound Exposure
Sample Preparation
Mass Spectrometry
Data Analysis

Cell Culture
- Expand iPS cells (HYR0103, ATCC) on Matrigel in mTeSR1
- Plate in 96-well assay plates at 100,000 cells/well

Compound Exposure
- Expose iPS cells to compound for 48 hours
- Collect spent media for LC-MS analysis
- Analyze viability

Sample Preparation
- Filter samples to remove molecules >10KDa

Mass Spectrometry
- Analyze spent media with UPLC-ESI-TOF-MS to determine levels of ornithine and cystine

Data Analysis
- Calculate fold change for each metabolite in treated samples relative to DMSO controls
- Determine o/c ratio
- Fit a 4 parameter log-logistic model
- Predict developmental toxicity potential

Study Design

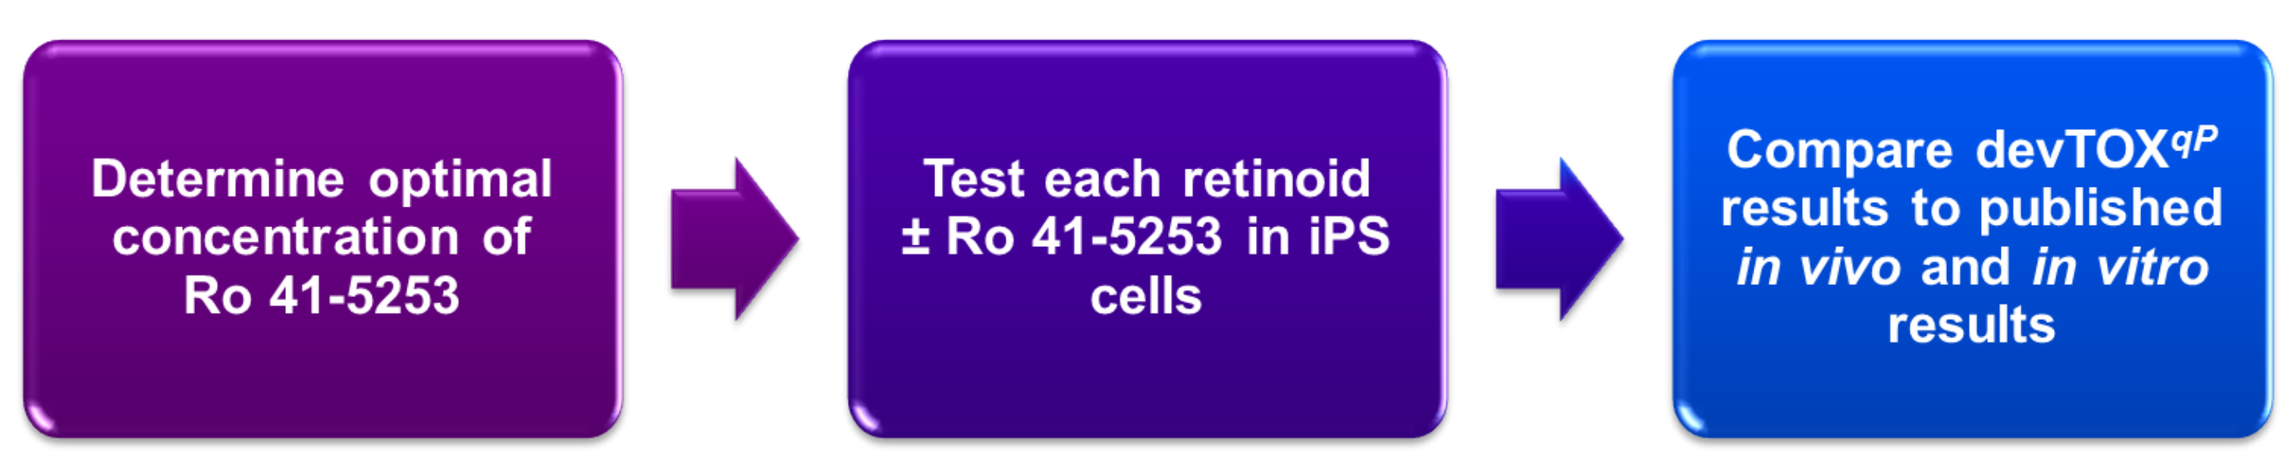
Determine optimal concentration of Ro 41-5253
Test each retinoid ± Ro 41-5253 in iPS cells
Compare devTOX^qP results to published in vivo and in vitro results

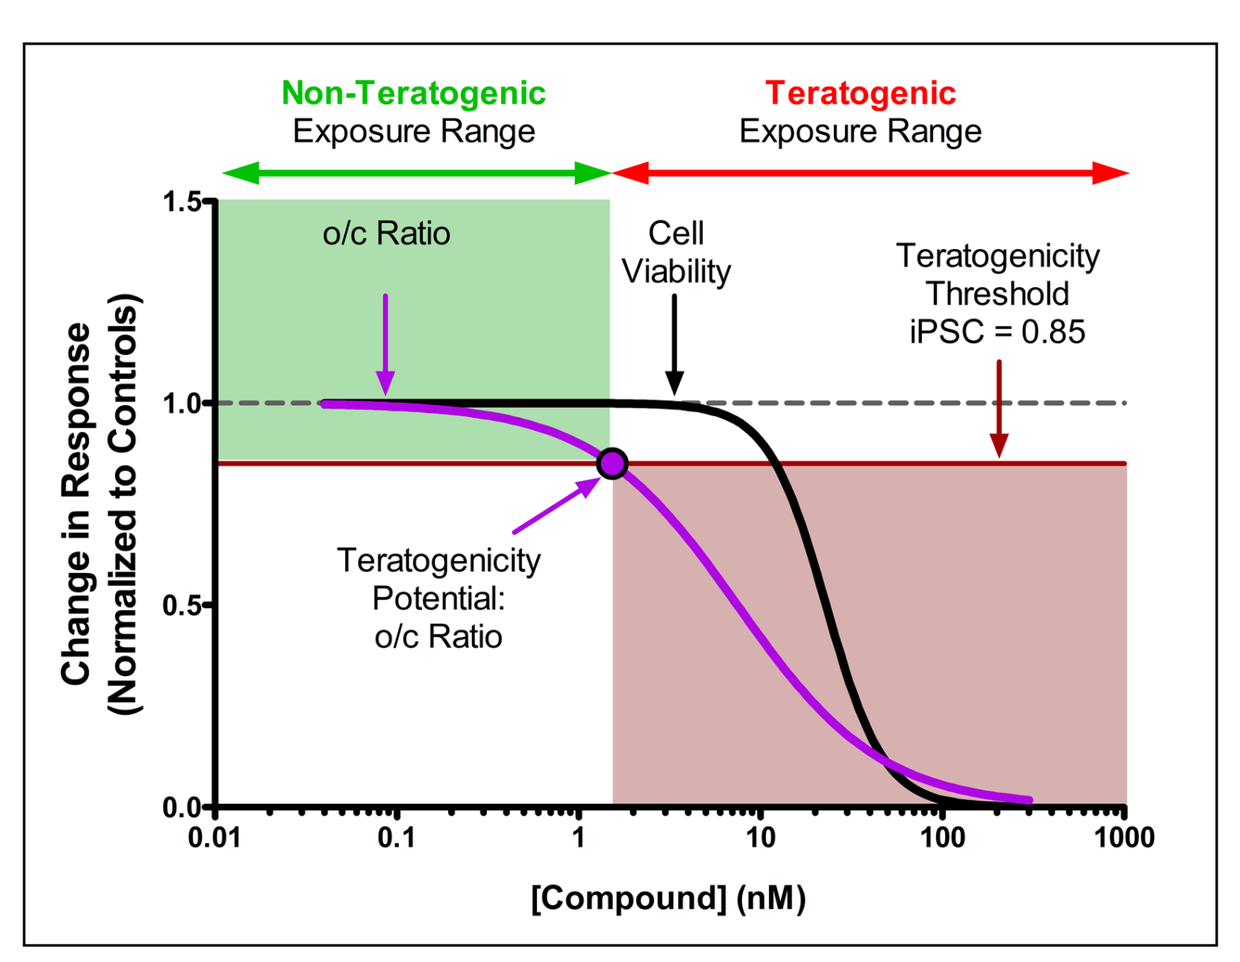
Non-Teratogenic
Exposure Range
Teratogenic
Exposure Range
o/c Ratio
Cell Viability
Teratogenicity Threshold iPSC = 0.85
Teratogenicity Potential: o/c Ratio
Change in Response (Normalized to Controls)
[Compound] (nM)

Graphical representation of devTOX^{qP} results.

Test Compounds

Test Compound	Synonyms	Preclinical *in vivo* and known human developmental effects[a]
13-*cis* Retinoic Acid	Isotretinoin Accutane	Craniofacial, central nervous system, cardiovascular, limb, skeletal
9-*cis* Retinoic Acid	Alitretinoin Panretin	Craniofacial, limb, embryo lethality
All-*trans* Retinoic Acid	Tretinoin Retin A	Craniofacial, central nervous system, cardiovascular, limb, skeletal, embryo/fetal lethality
Etretinate	Tegison	Craniofacial, central nervous system, cardiovascular, limb, skeletal, embryo/fetal lethality
Acitretin[b]	Soriatane	Craniofacial, central nervous system, cardiovascular, limb, skeletal, embryo/fetal lethality
Retinol	Vitamin A	Low doses: none High doses: craniofacial, central nervous system, cardiovascular, skeletal
TTNPB	Arotinoid acid Ro 13-7410	Craniofacial, central nervous system, cardiovascular, limb, skeletal, embryo/fetal lethality

[a] The preclinical *in vivo* and known human developmental effects were summarized from the Teratogen Information System (TERIS) or the compound's FDA label.

[b] Acitretin is the active metabolite of Etretinate.

Test Compound Structures and Relationship

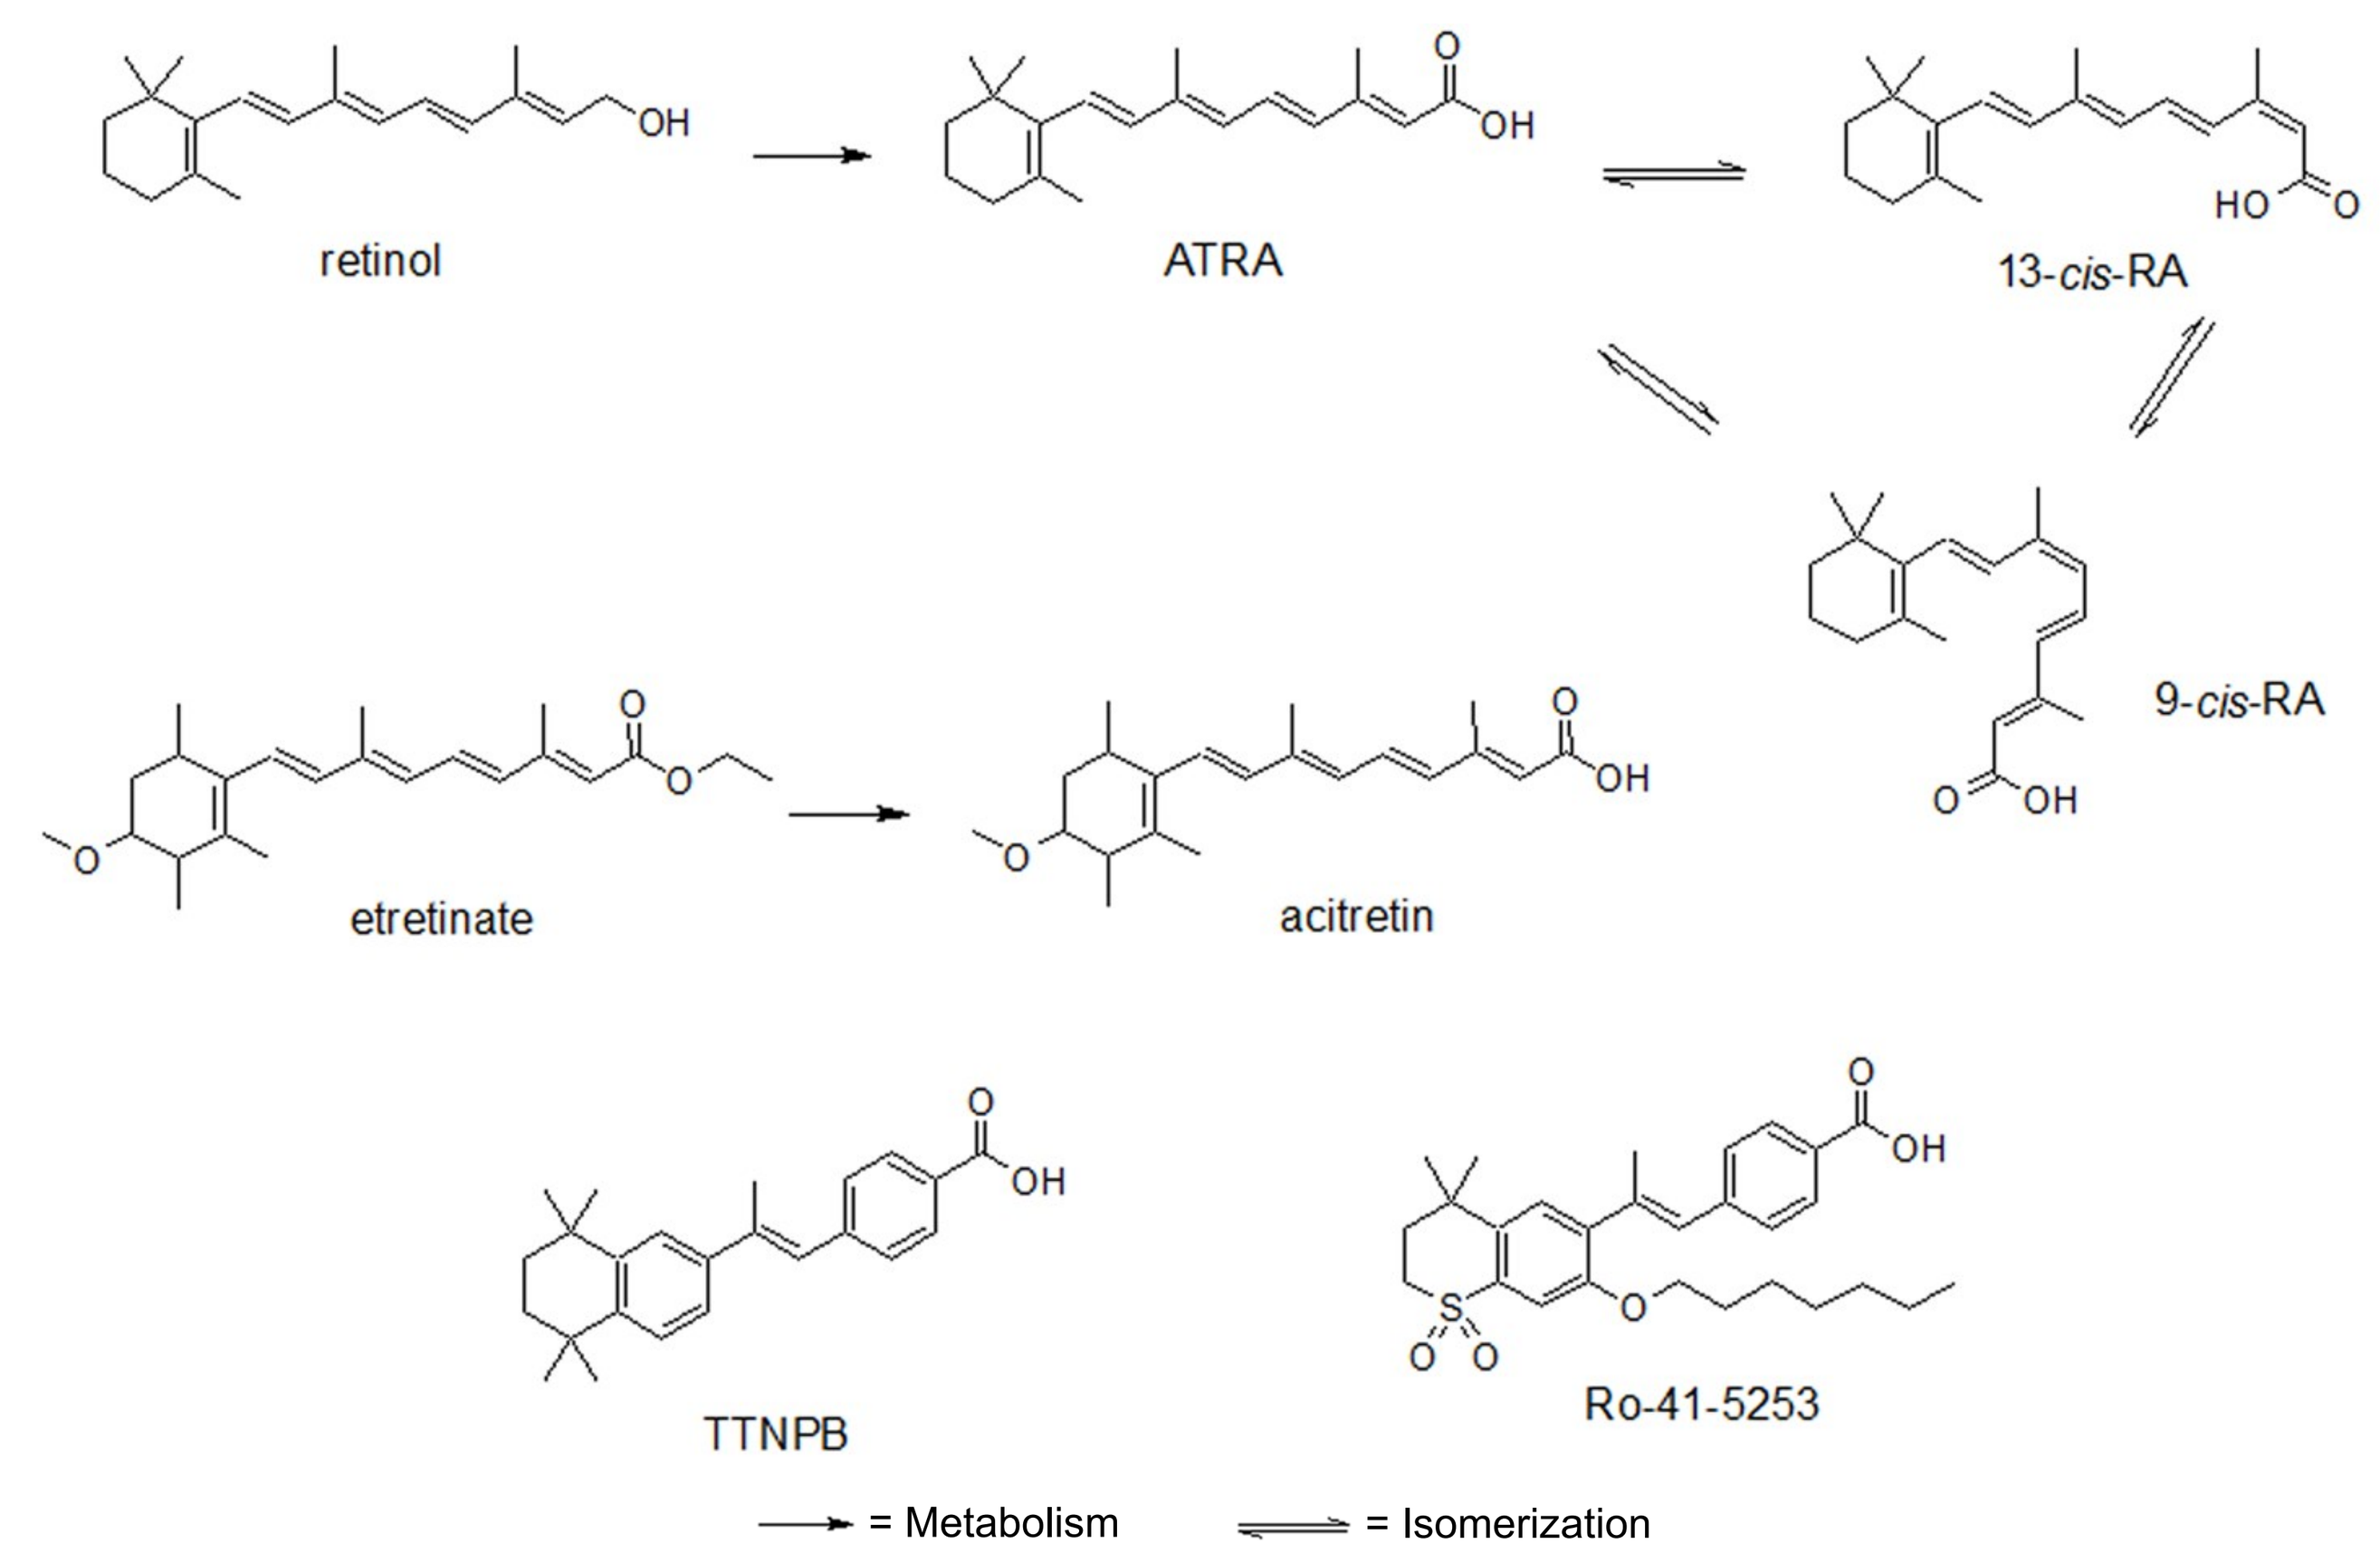
retinol
ATRA
13-cis-RA
9-cis-RA
etretinate
acitretin
TTNPB
Ro-41-5253
= Metabolism
= Isomerization

Results

6 µM Ro 41-5253 Inhibits 10 nM All-*trans* Retinoic Acid-Induced Response in the o/c Ratio in iPS Cells

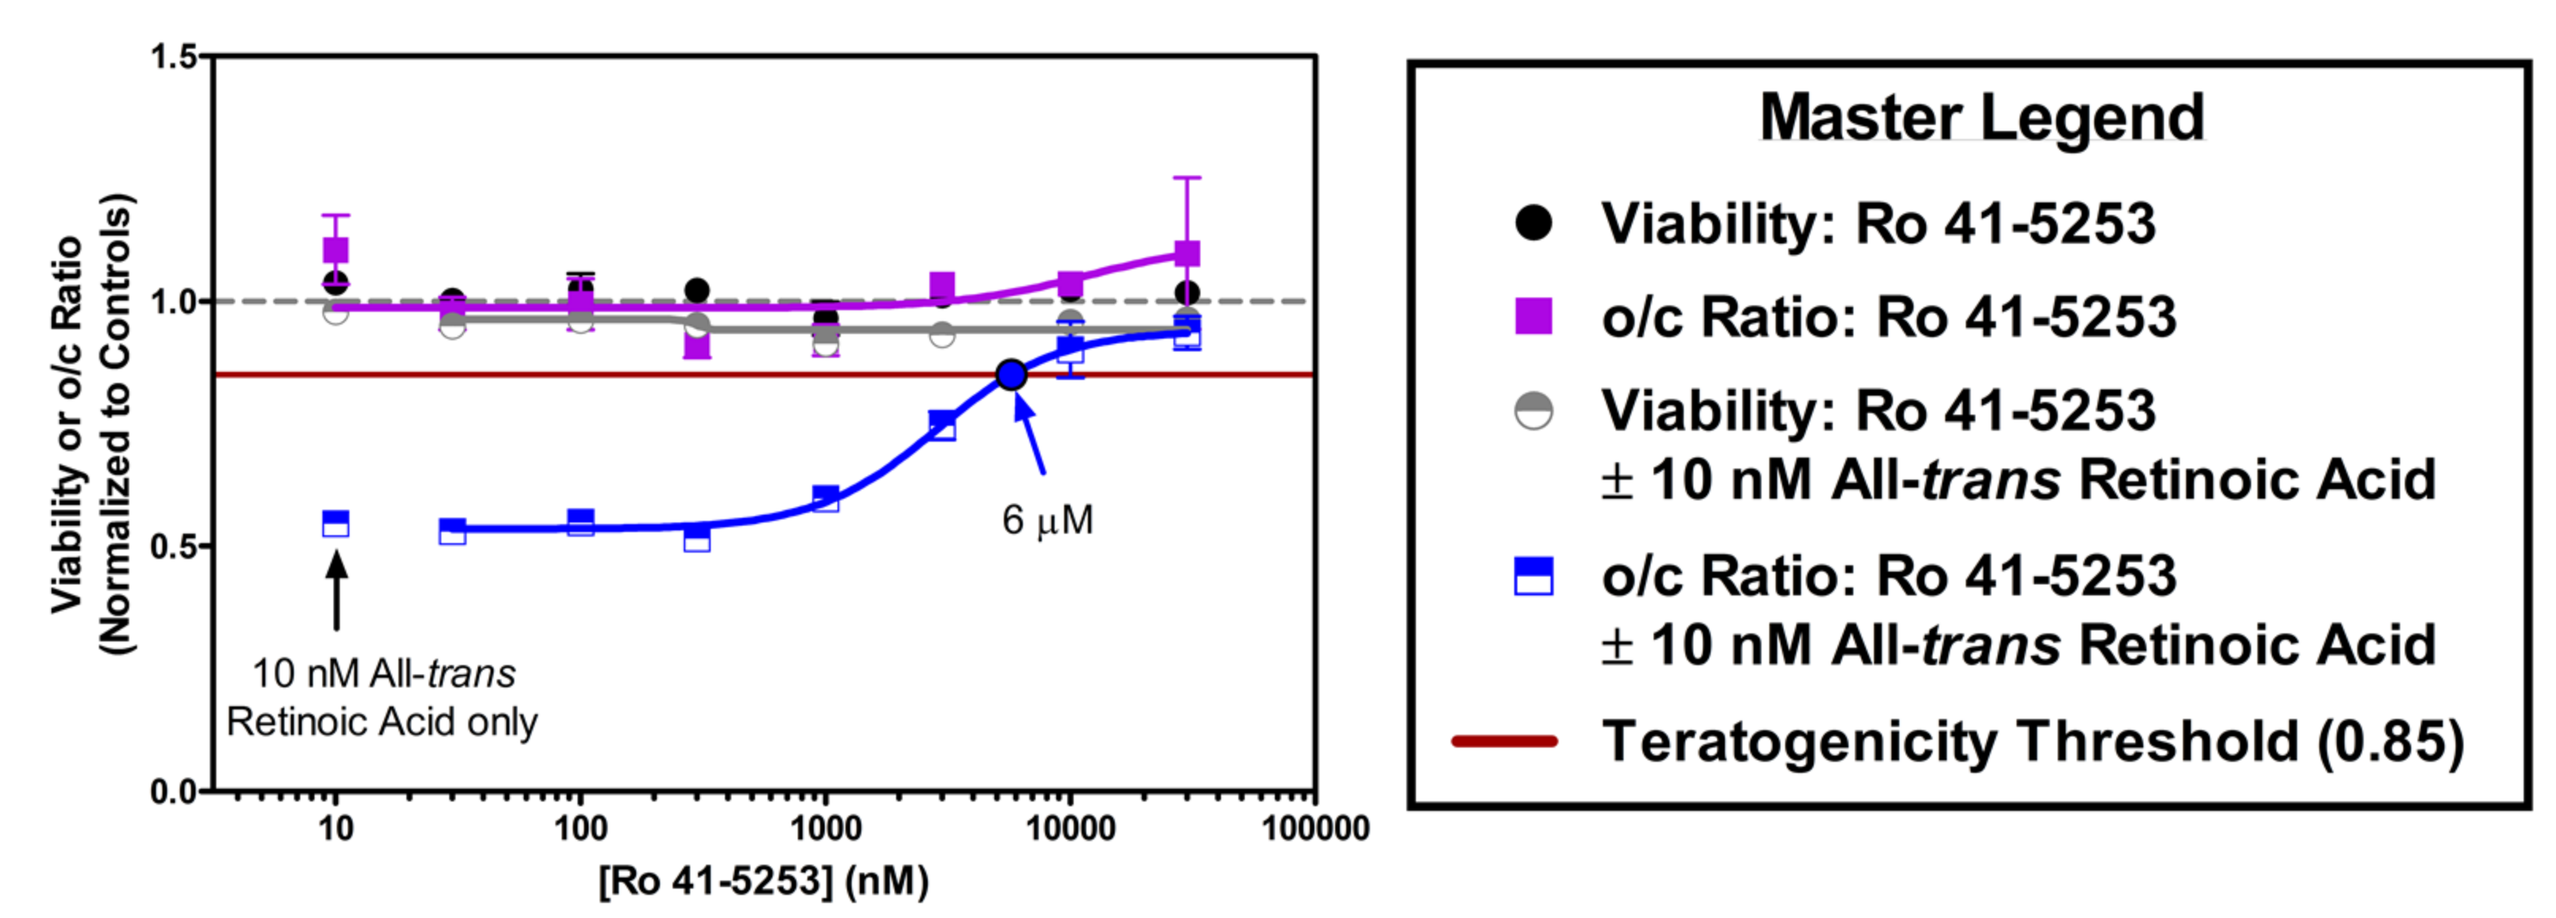
Viability or o/c Ratio (Normalized to Controls)
[Ro 41-5253] (nM)
10 nM All-trans Retinoic Acid only
6 µM
Master Legend
Viability: Ro 41-5253
o/c Ratio: Ro 41-5253
Viability: Ro 41-5253 ± 10 nM All-trans Retinoic Acid
o/c Ratio: Ro 41-5253 ± 10 nM All-trans Retinoic Acid
Teratogenicity Threshold (0.85)

- 10 µM Ro 41-5253 was chosen for antagonist exposure experiments.

iPS cells Exhibit a Biphasic Response to All-*trans* Retinoic Acid

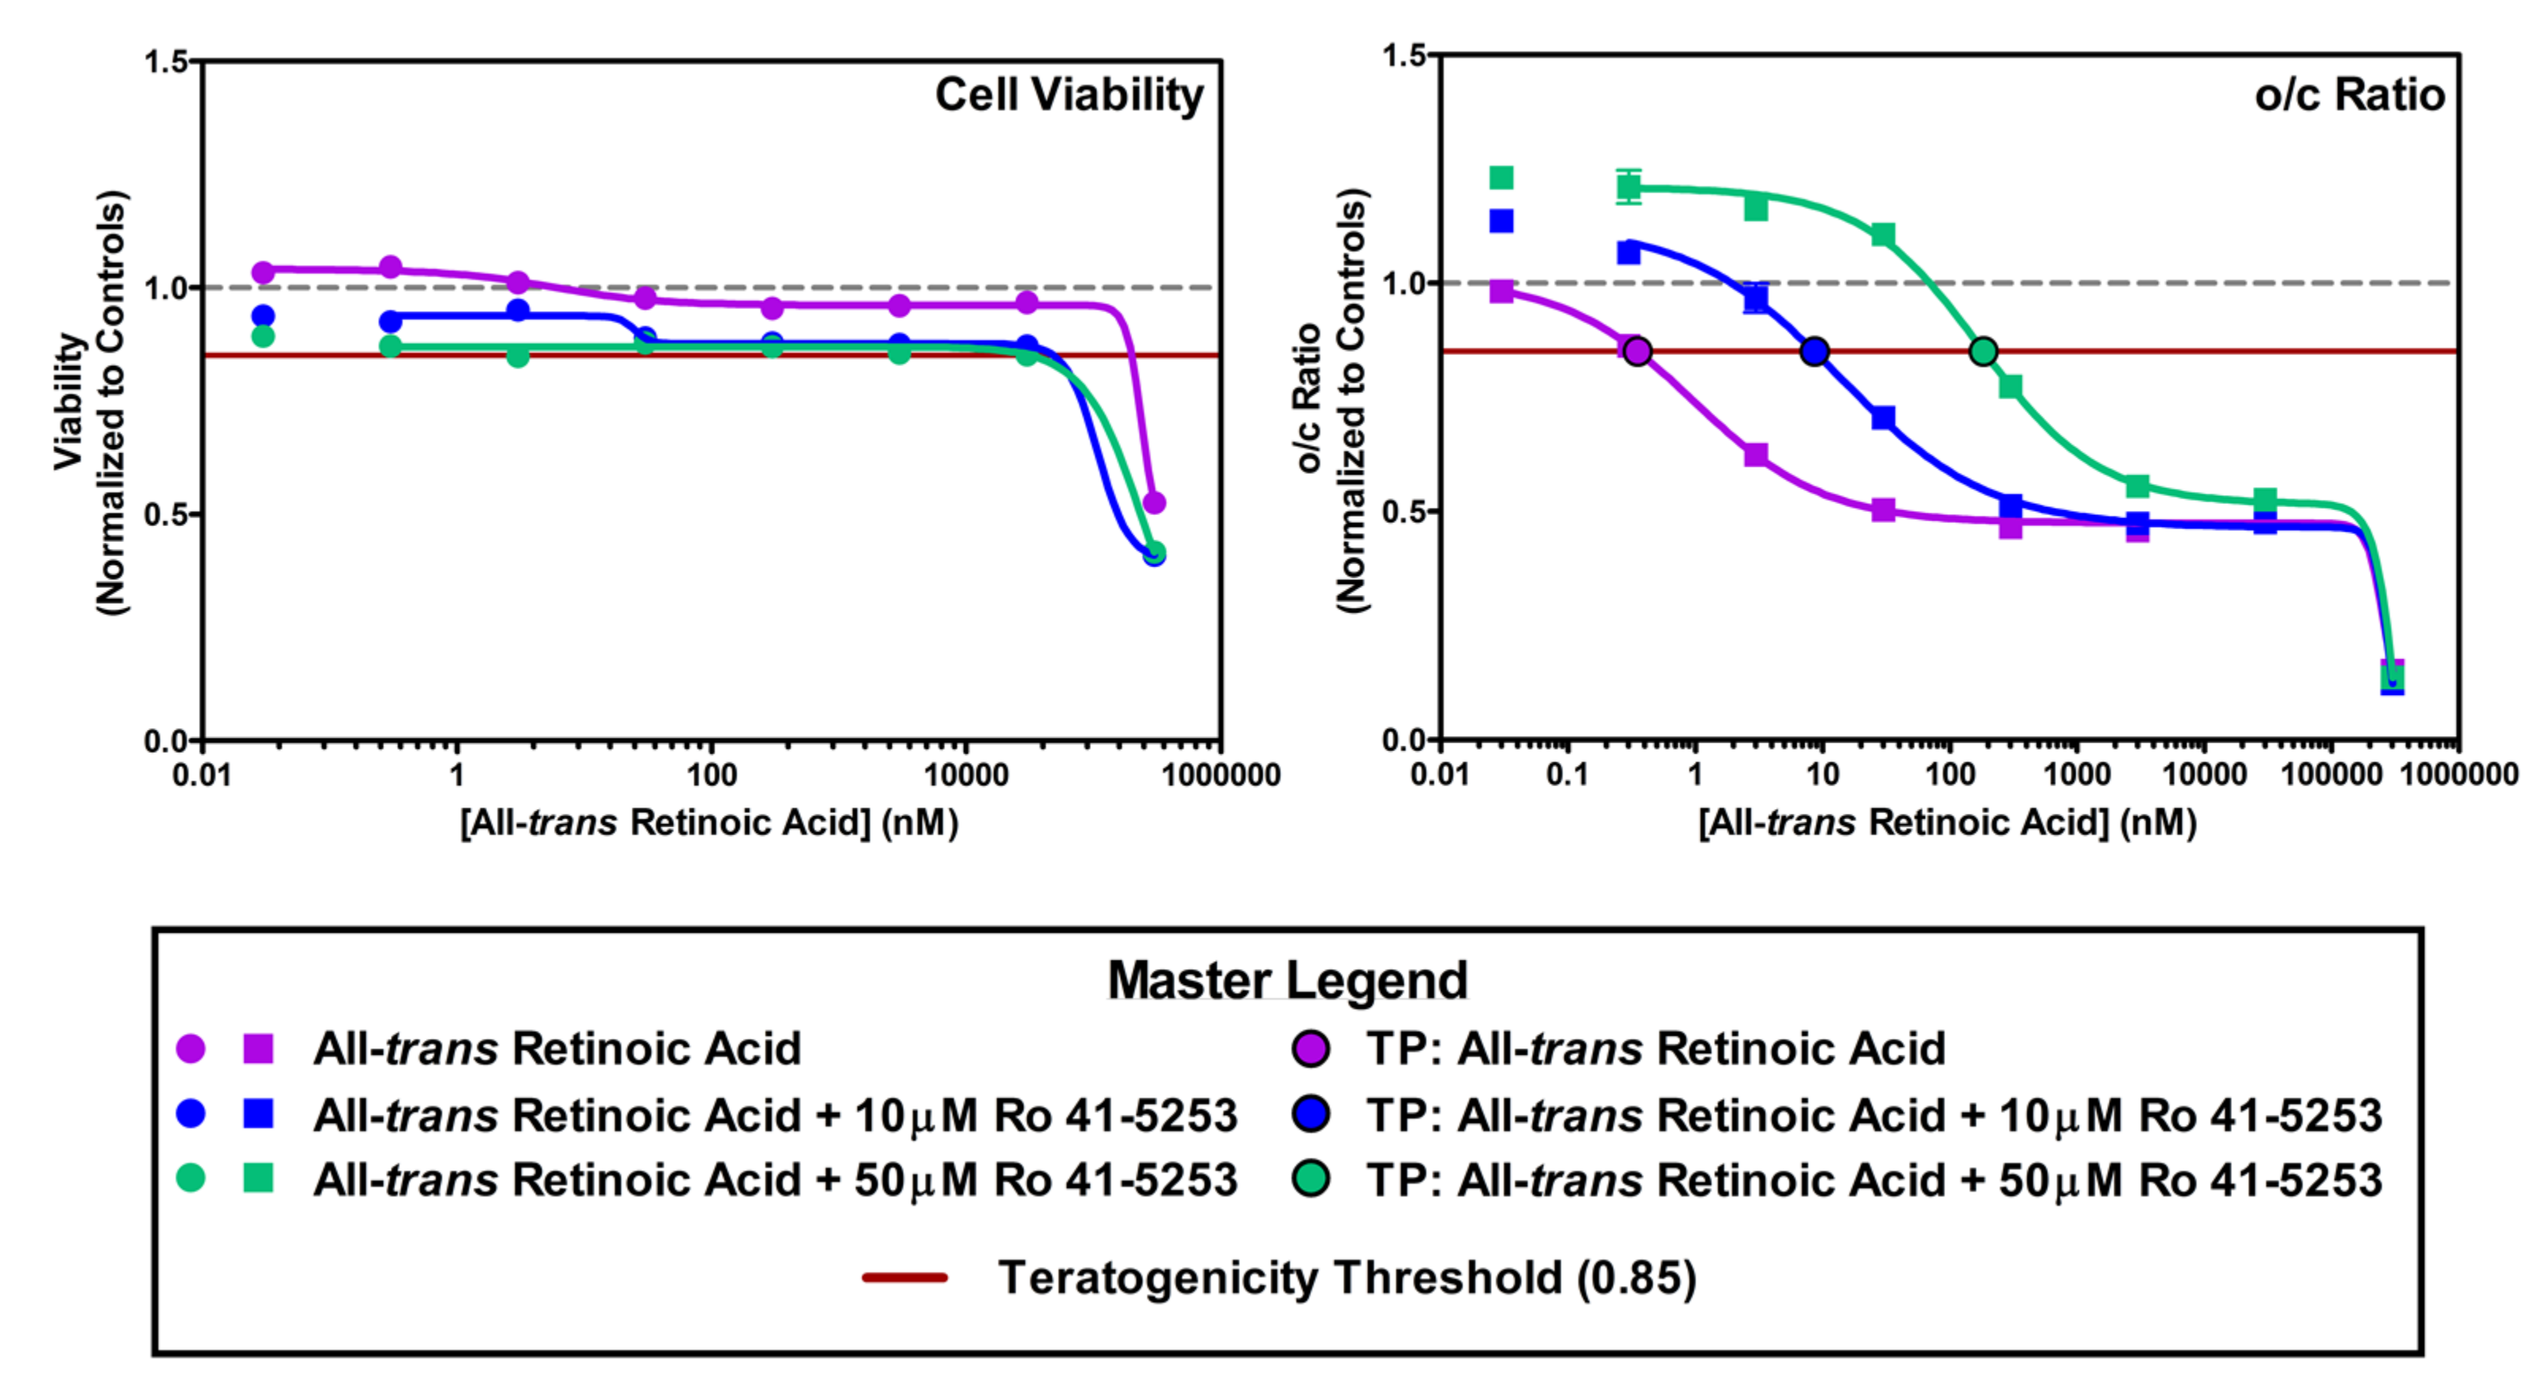
Cell Viability
o/c Ratio
Viability (Normalized to Controls)
o/c Ratio (Normalized to Controls)
[All-trans Retinoic Acid] (nM)
Master Legend
All-trans Retinoic Acid
All-trans Retinoic Acid + 10µM Ro 41-5253
All-trans Retinoic Acid + 50µM Ro 41-5253
TP: All-trans Retinoic Acid
TP: All-trans Retinoic Acid + 10µM Ro 41-5253
TP: All-trans Retinoic Acid + 50µM Ro 41-5253
Teratogenicity Threshold (0.85)

	All-*trans* Retinoic Acid	All-*trans* Retinoic Acid + 10µM Ro 41-5253	All-*trans* Retinoic Acid + 50µM Ro 41-5253
Teratogenicity Potential (nM)	0.35	8.6	185

- The metabolic response elicited by all-*trans* retinoic acid is independent of changes in cell viability.
- Ro 41-5253 counteracts the effects of all-*trans* retinoic acid on metabolism but not changes in cell viability.

Developmental Toxicity Response is Mediated through the Retinoic Acid Receptor

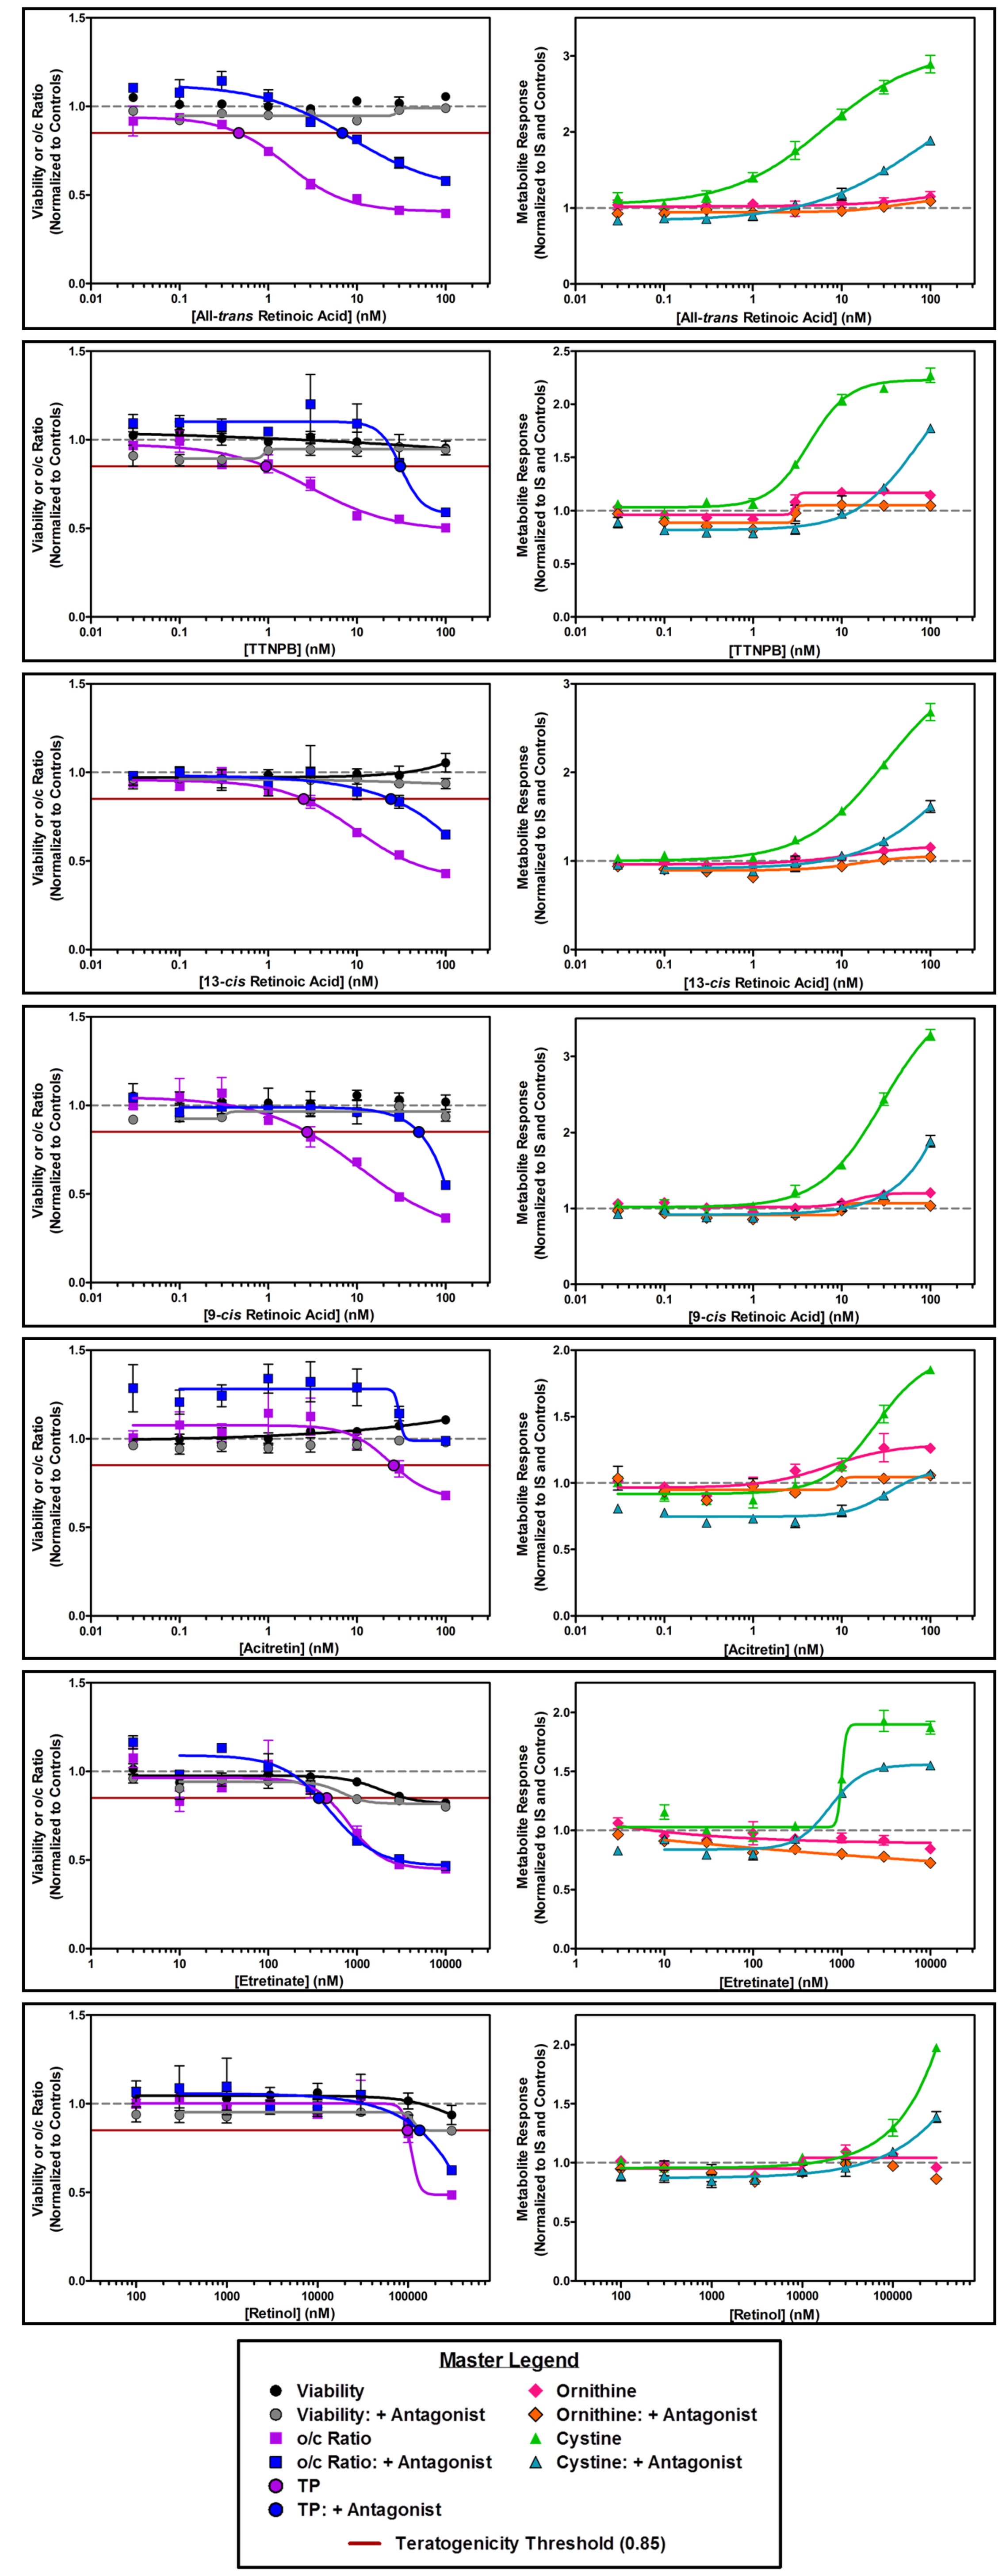
Viability or o/c Ratio (Normalized to Controls)
Metabolite Response (Normalized to IS and Controls)
[All-trans Retinoic Acid] (nM)
[TTNPB] (nM)
[13-cis Retinoic Acid] (nM)
[9-cis Retinoic Acid] (nM)
[Acitretin] (nM)
[Etretinate] (nM)
[Retinol] (nM)
Master Legend
Viability
Viability: + Antagonist
o/c Ratio
o/c Ratio: + Antagonist
TP
TP: + Antagonist
Ornithine
Ornithine: + Antagonist
Cystine
Cystine: + Antagonist
Teratogenicity Threshold (0.85)

Representative results for each compound tested. Data was taken from the same replicate for each compound.

devTOX^{qP} Results Match Published *In Vitro* Results but Differ Slightly from Published *In Vivo* Results

Test Compound	iPSC devTOX^{qP} Results		mEST BMC_{50} (nM)[b]
	- Antagonist (nM)[a]	+ Antagonist (nM)[a]	
All-*trans* Retinoic Acid	0.49 (±0.63)	14 (±22)	3.3 (±1.8)
TTNPB	0.98 (±1.1)	38 (±49)	0.17 (±0.026)
13-*cis* Retinoic Acid	2.0 (±1.5)	47 (±35)	5.0 (±3.3)
9-*cis* Retinoic Acid	4.1 (±5.9)	55 (±32)	10 (±8.6)
Acitretin	18 (±19)	96 (±7.5)	12 (±6.3)
Etretinate	1227 (±1633)	1832 (±3109)	1500 (±610)
Retinol	118565 (±127850)	184099 (±83991)	2800 (±1200)

[a] Values are average interpolated teratogenicity potential concentration (±SD). Note: if the compound did not exhibit a response, the highest exposure of the dose range was used to calculate the average.

[b] From Louisse et al., 2011. BMC_{50}: benchmark concentration at which a benchmark response of 50% is reached. Value is average of 3 replicates (±SD).

- iPS cells exhibited differing teratogenicity potential concentrations.
- Ro 41-5253 inhibited metabolic perturbation indicating that the teratogenic response is mediated through the retinoic acid receptor.
- devTOX^{qP} teratogenicity potential concentrations were highly correlated with mEST BMC_{50} values (R^2=0.84, p-value=0.0036).

Test Compound	iPSC devTOX^{qP} Ranking	mEST Ranking[1]	*In Vivo* Ranking[a]				
			A	B	C	D	E
All-*trans* Retinoic Acid	1	2	2	1	1		3
TTNPB	2	1	1			1	1
13-*cis* Retinoic Acid	3	3			4	3	4
9-*cis* Retinoic Acid	4	4			3		
Acitretin	5	5		2		2	2
Etretinate	6	6			2		
Retinol	7	7	3				

[a] From Louisse et al., 2011. *In vivo* rankings dependent on species and/or exposure regimen used. **A**: Rats exposed on GD 9 or 10. **B**: Rats exposed on GD10 or 11. **C**: Mice exposed on GD11. **D**: Mice exposed on GD7-16. **E**: Rabbits exposed on GD7-19 or GD 6-18.

- All-*trans* Retinoic Acid and TTNPB were the most potent *in vivo* and in iPS cells.
- Retinol was the least potent *in vivo* and in iPS cells.
- Acitretin and Etretinate were more potent than 13-*cis* and 9-*cis* Retinoic Acid *in vivo* but less potent *in vitro*.

Conclusions

- The current study demonstrates the utility of this human cell-based assay toward ranking compound series.
- Compound ranking trends are concordant with *in vivo* and other *in vitro* models.
- Use of the antagonist demonstrates that the developmental toxicity response is mediated via the RAR pathway.
- Changes in the metabolic response are independent of cell viability, with changes in cell viability observed at only at very high exposures.
- iPS cells do not have the ability to metabolize etretinate to its active form.
- devTOX^{qP} is a relevant screen for developmental toxicity that can be applied for ranking compound series with a high degree of concordance with current validated *in vitro* and *in vivo* models.

References

Louisse J, et al. Relative developmental toxicity potencies of retinoids in the embryonic stem cell test compared with their relative potencies in *in vivo* and two other *in vitro* assays for developmental toxicity. *Toxicology Letters*. 2011; **203**:1-8.